Safety and Efficacy of KarXT (Xanomeline–Trospium) in Patients With Schizophrenia: Results From a Phase 3, Randomised, Double-Blind, Placebo-Controlled Trial (EMERGENT-2)

Christoph U. Correll,[1-3] Angel S. Angelov,[4] Stephen K. Brannan[4]

[1]The Zucker Hillside Hospital, Glen Oaks, NY, USA; [2]Donald and Barbara Zucker School of Medicine at Hofstra/Northwell, Hempstead, NY, USA; [3]Charité Universitätsmedizin Berlin, Berlin, Germany; [4]Karuna Therapeutics, Boston, MA, USA

ECNP Congress
P.0193

Background

- KarXT combines the dual M_1/M_4 preferring muscarinic receptor agonist xanomeline with the peripherally restricted muscarinic receptor antagonist trospium
- In the 5-week, randomised, double-blind, placebo-controlled, phase 2 EMERGENT-1 trial (NCT03697252), KarXT met the primary endpoint of a significant reduction in Positive and Negative Syndrome Scale (PANSS) total score through week 5 vs placebo, improved other key efficacy measures, and was generally well tolerated[1]

Methods

- EMERGENT-2 (NCT04659161) was a phase 3, randomised, double-blind, placebo-controlled, 5-week trial of KarXT vs placebo (**Figure 1**)

Figure 1. EMERGENT-2 Trial Design

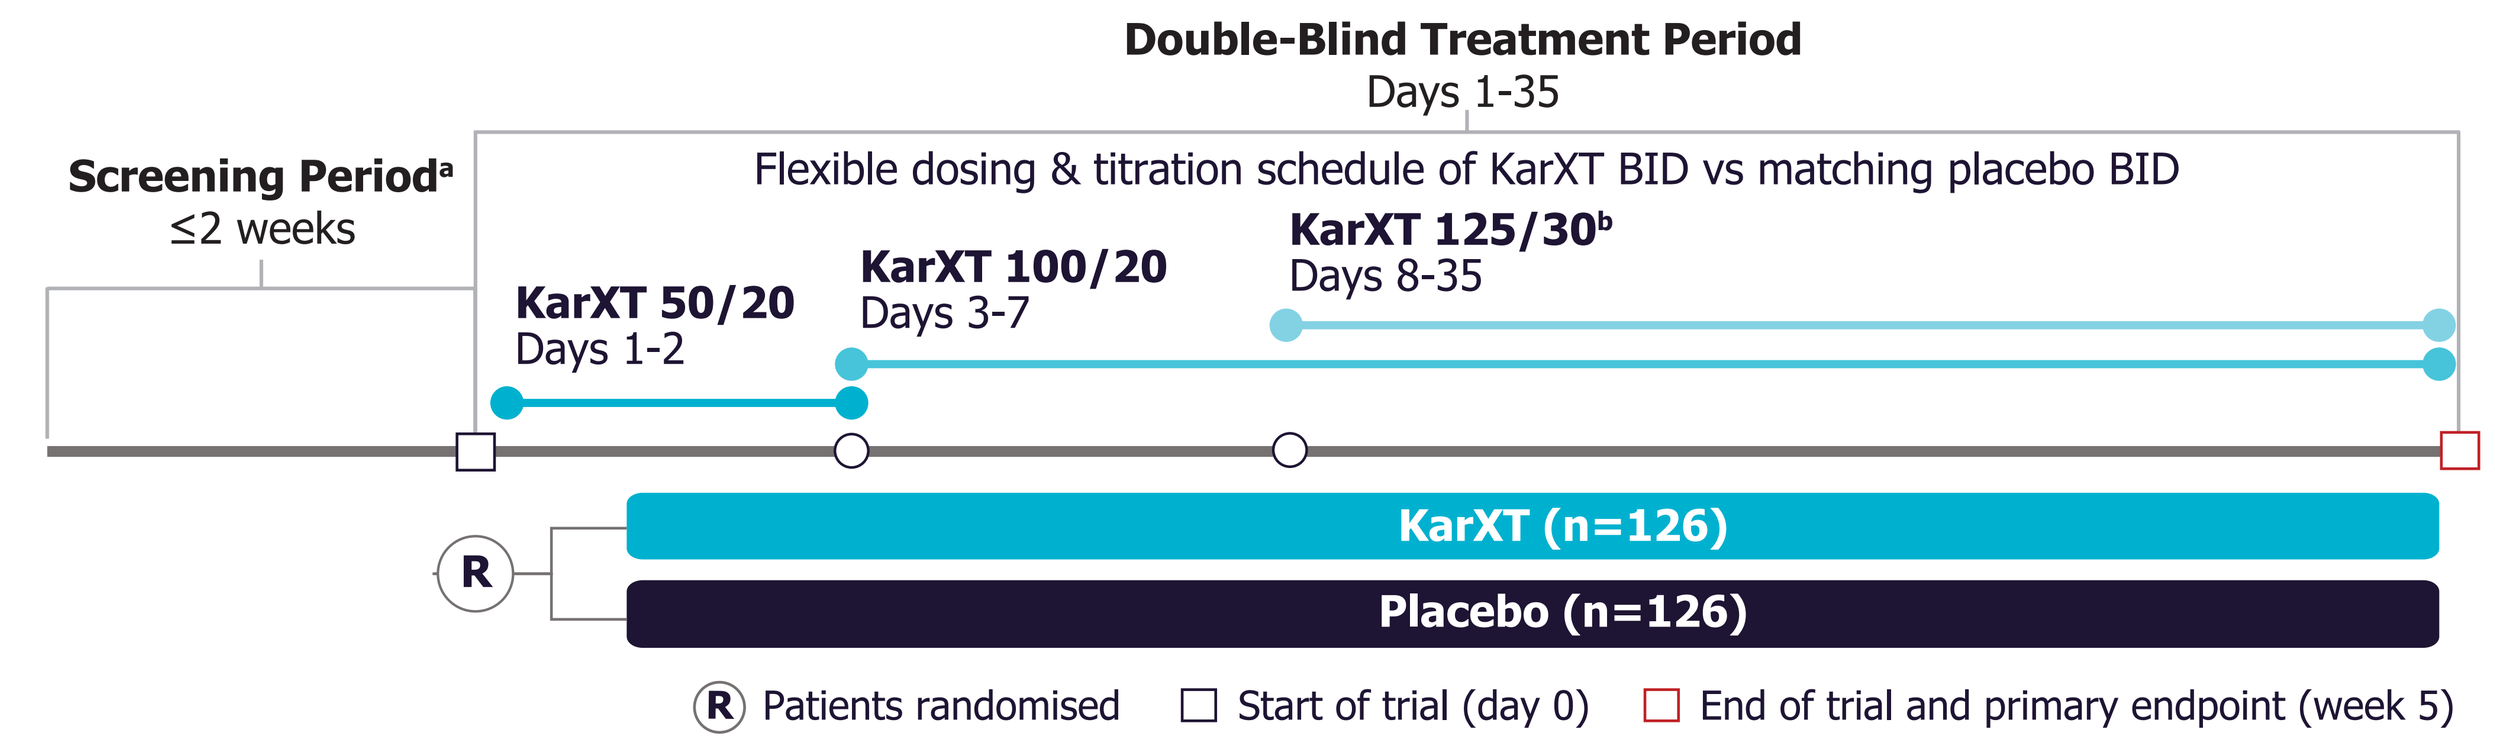
Double-Blind Treatment Period
Days 1-35
Screening Period[a]
≤2 weeks
Flexible dosing & titration schedule of KarXT BID vs matching placebo BID
KarXT 50/20
Days 1-2
KarXT 100/20
Days 3-7
KarXT 125/30[b]
Days 8-35
R
KarXT (n=126)
Placebo (n=126)
R Patients randomised
Start of trial (day 0)
End of trial and primary endpoint (week 5)

KarXT dose is expressed as xanomeline/trospium (mg/mg). [a]Washout of prior oral lithium and/or antipsychotics. [b]Optional increase in dose based on tolerability determined by a clinician. BID, twice daily.

- Adult patients aged 18-65 years with a confirmed *DSM-5* diagnosis of schizophrenia and a recent worsening of psychotic symptoms warranting hospitalisation were enrolled
- Eligible patients were randomised 1:1 to KarXT or matched placebo
- Dosing of KarXT (xanomeline/trospium) started at 50 mg/20 mg twice daily (BID) and increased to a maximum dose of 125 mg/30 mg BID
- Primary efficacy endpoint: change from baseline to week 5 in PANSS total score compared with placebo
- Secondary efficacy endpoints: change from baseline to week 5 in PANSS positive subscale, PANSS negative subscale, and PANSS Marder negative factor subscale scores; Clinical Global Impression–Severity at week 5[a]; and percentage of PANSS responders at week 5[a]

Statistical Analyses

- Efficacy analyses were performed in the modified intent-to-treat population, defined as all randomised patients who received ≥1 dose of study medication, had a baseline PANSS assessment, and had ≥1 postbaseline PANSS assessment
- Safety analyses were performed in the safety population, defined as all patients who received ≥1 dose of study drug

[a]Analysis is ongoing and results to be presented at a future meeting.

Results

- A total of 252 patients at 22 study sites in the United States were enrolled
- There were no meaningful differences in baseline demographics and characteristics between treatment groups (**Table 1**)

Table 1. Baseline Demographics and Characteristics (ITT Population)

	KarXT (n=126)	Placebo (n=126)
Mean age, years (SD)	45.6 (10.4)	46.2 (10.8)
Sex, n (%)		
Male	95 (75.4)	95 (75.4)
Female	31 (24.6)	31 (24.6)
Race, n (%)		
Asian	2 (1.6)	1 (0.8)
Black	97 (77.0)	92 (73.0)
White	26 (20.6)	31 (24.6)
Other	1 (0.8)	2 (1.6)
PANSS total score, mean (SD)	98.3 (8.9)	97.9 (9.7)
PANSS positive subscale score, mean (SD)	26.8 (3.7)	26.7 (4.0)
PANSS negative subscale score, mean (SD)	22.9 (4.0)	22.9 (3.8)
PANNS Marder negative factor subscale score, mean (SD)	22.9 (5.0)	22.5 (4.7)

ITT defined as all randomised patients.
ITT, intent-to-treat; PANSS, Positive and Negative Syndrome Scale.

Primary Endpoint: Change in PANSS Total Score vs Placebo at Week 5

- KarXT demonstrated a statistically significant and clinically meaningful 9.6-point reduction in PANSS total score compared with placebo at week 5 (-21.2 KarXT vs -11.6 placebo, *P*<0.0001; Cohen's d effect size=0.61) (**Figure 2**)
- KarXT demonstrated a statistically significant improvement in PANSS total score starting at week 2 (first postbaseline rating) and maintained such improvement through all time points in the trial

Figure 2. Change From Baseline in PANSS Total Score vs Placebo at Week 5

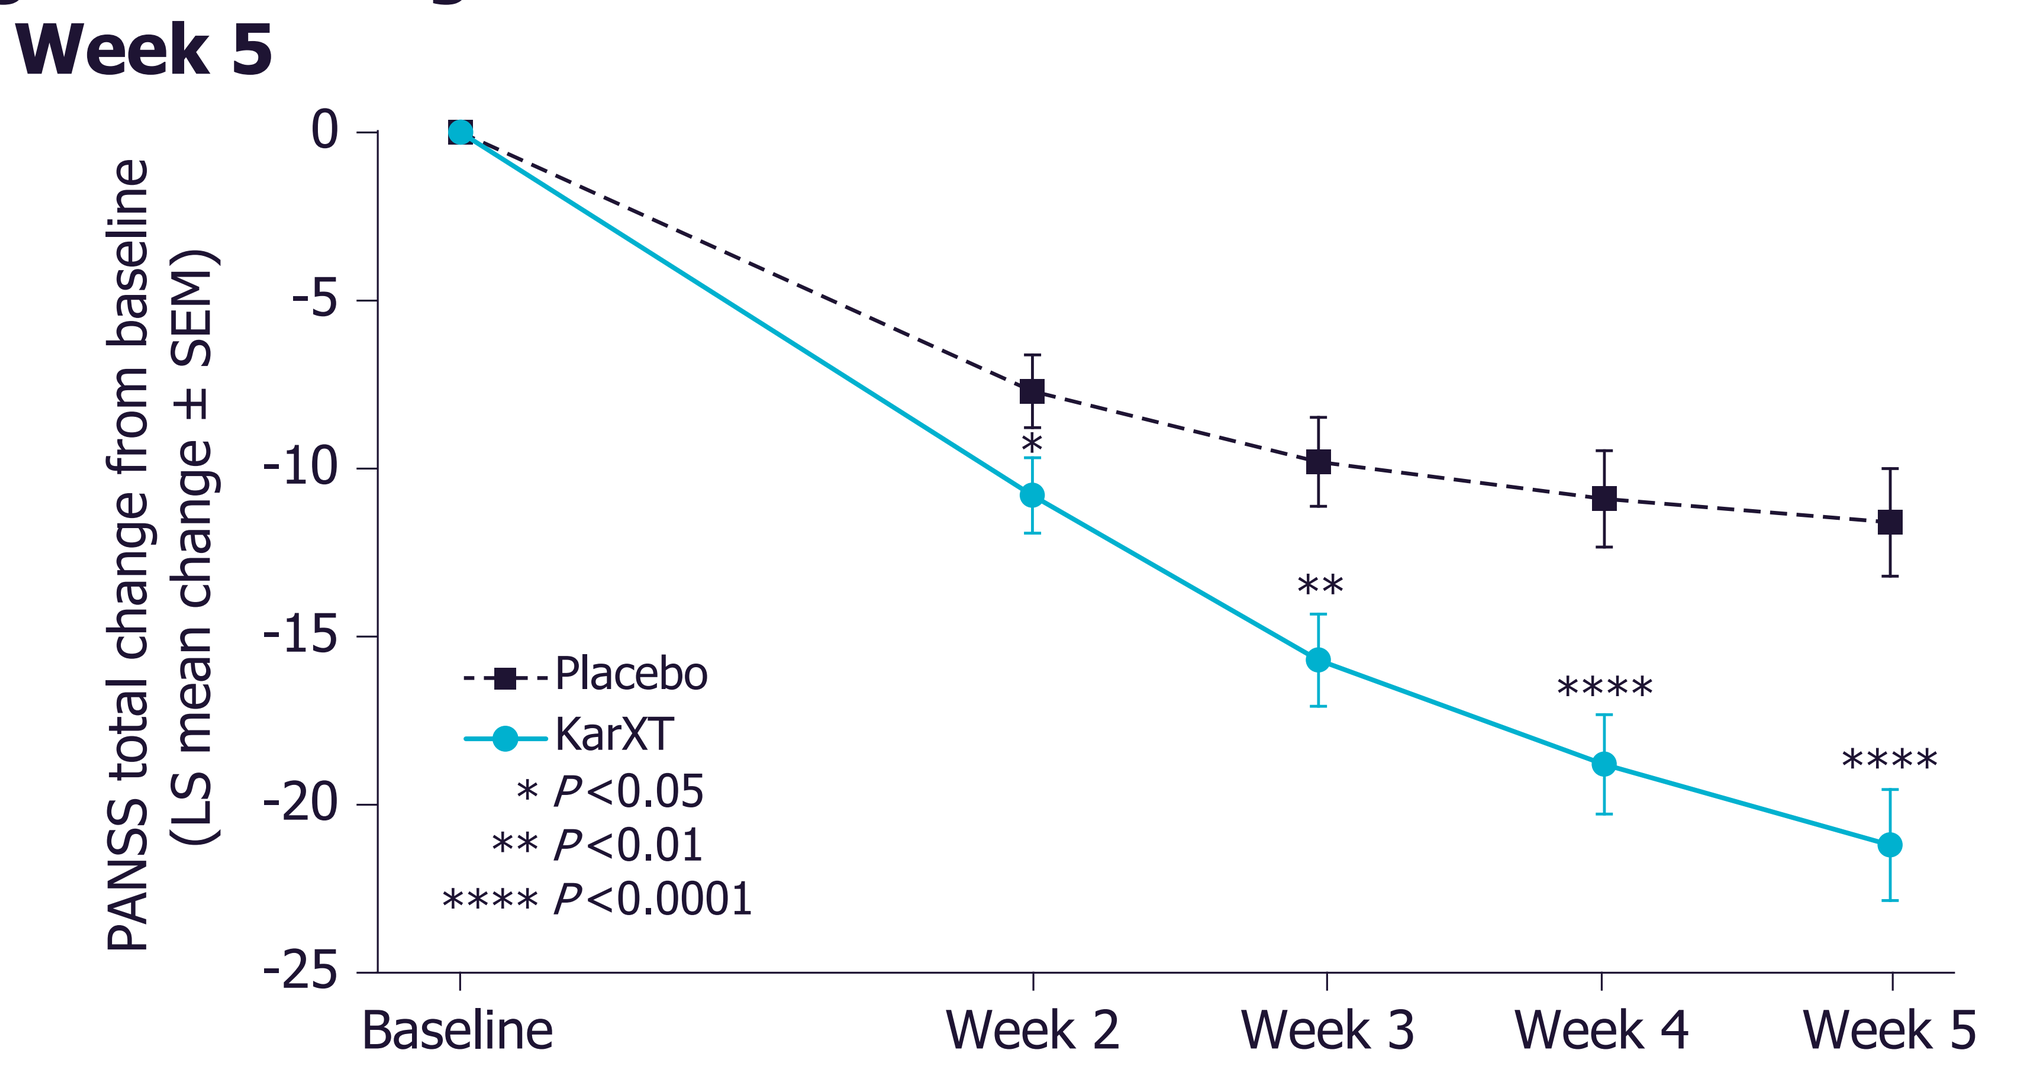
PANSS total change from baseline (LS mean change ± SEM)
0
-5
-10
-15
-20
-25
Baseline
Week 2
Week 3
Week 4
Week 5
Placebo
KarXT
* P<0.05
** P<0.01
**** P<0.0001

LS, least squares; PANSS, Positive and Negative Syndrome Scale; SEM, standard error of the mean.

Secondary Endpoints: Change in PANSS Subscale Scores vs Placebo at Week 5

- KarXT met tested secondary endpoints, demonstrating a statistically significant reduction in both positive and negative symptoms compared with placebo (**Figure 3**)

Safety and Tolerability

- KarXT was generally well tolerated (**Table 2**), with a side-effect profile substantially consistent with prior trials
- Overall discontinuation rates were similar between KarXT and placebo arms (25% vs 21%)
- Common treatment-emergent adverse events (TEAEs; ≥5%) were all mild to moderate in severity and mostly transient in nature

Figure 3. Change From Baseline in (A) PANSS Positive Subscale Score, (B) PANSS Negative Subscale Score, and (C) PANSS Marder Negative Factor Subscale Score vs Placebo at Week 5

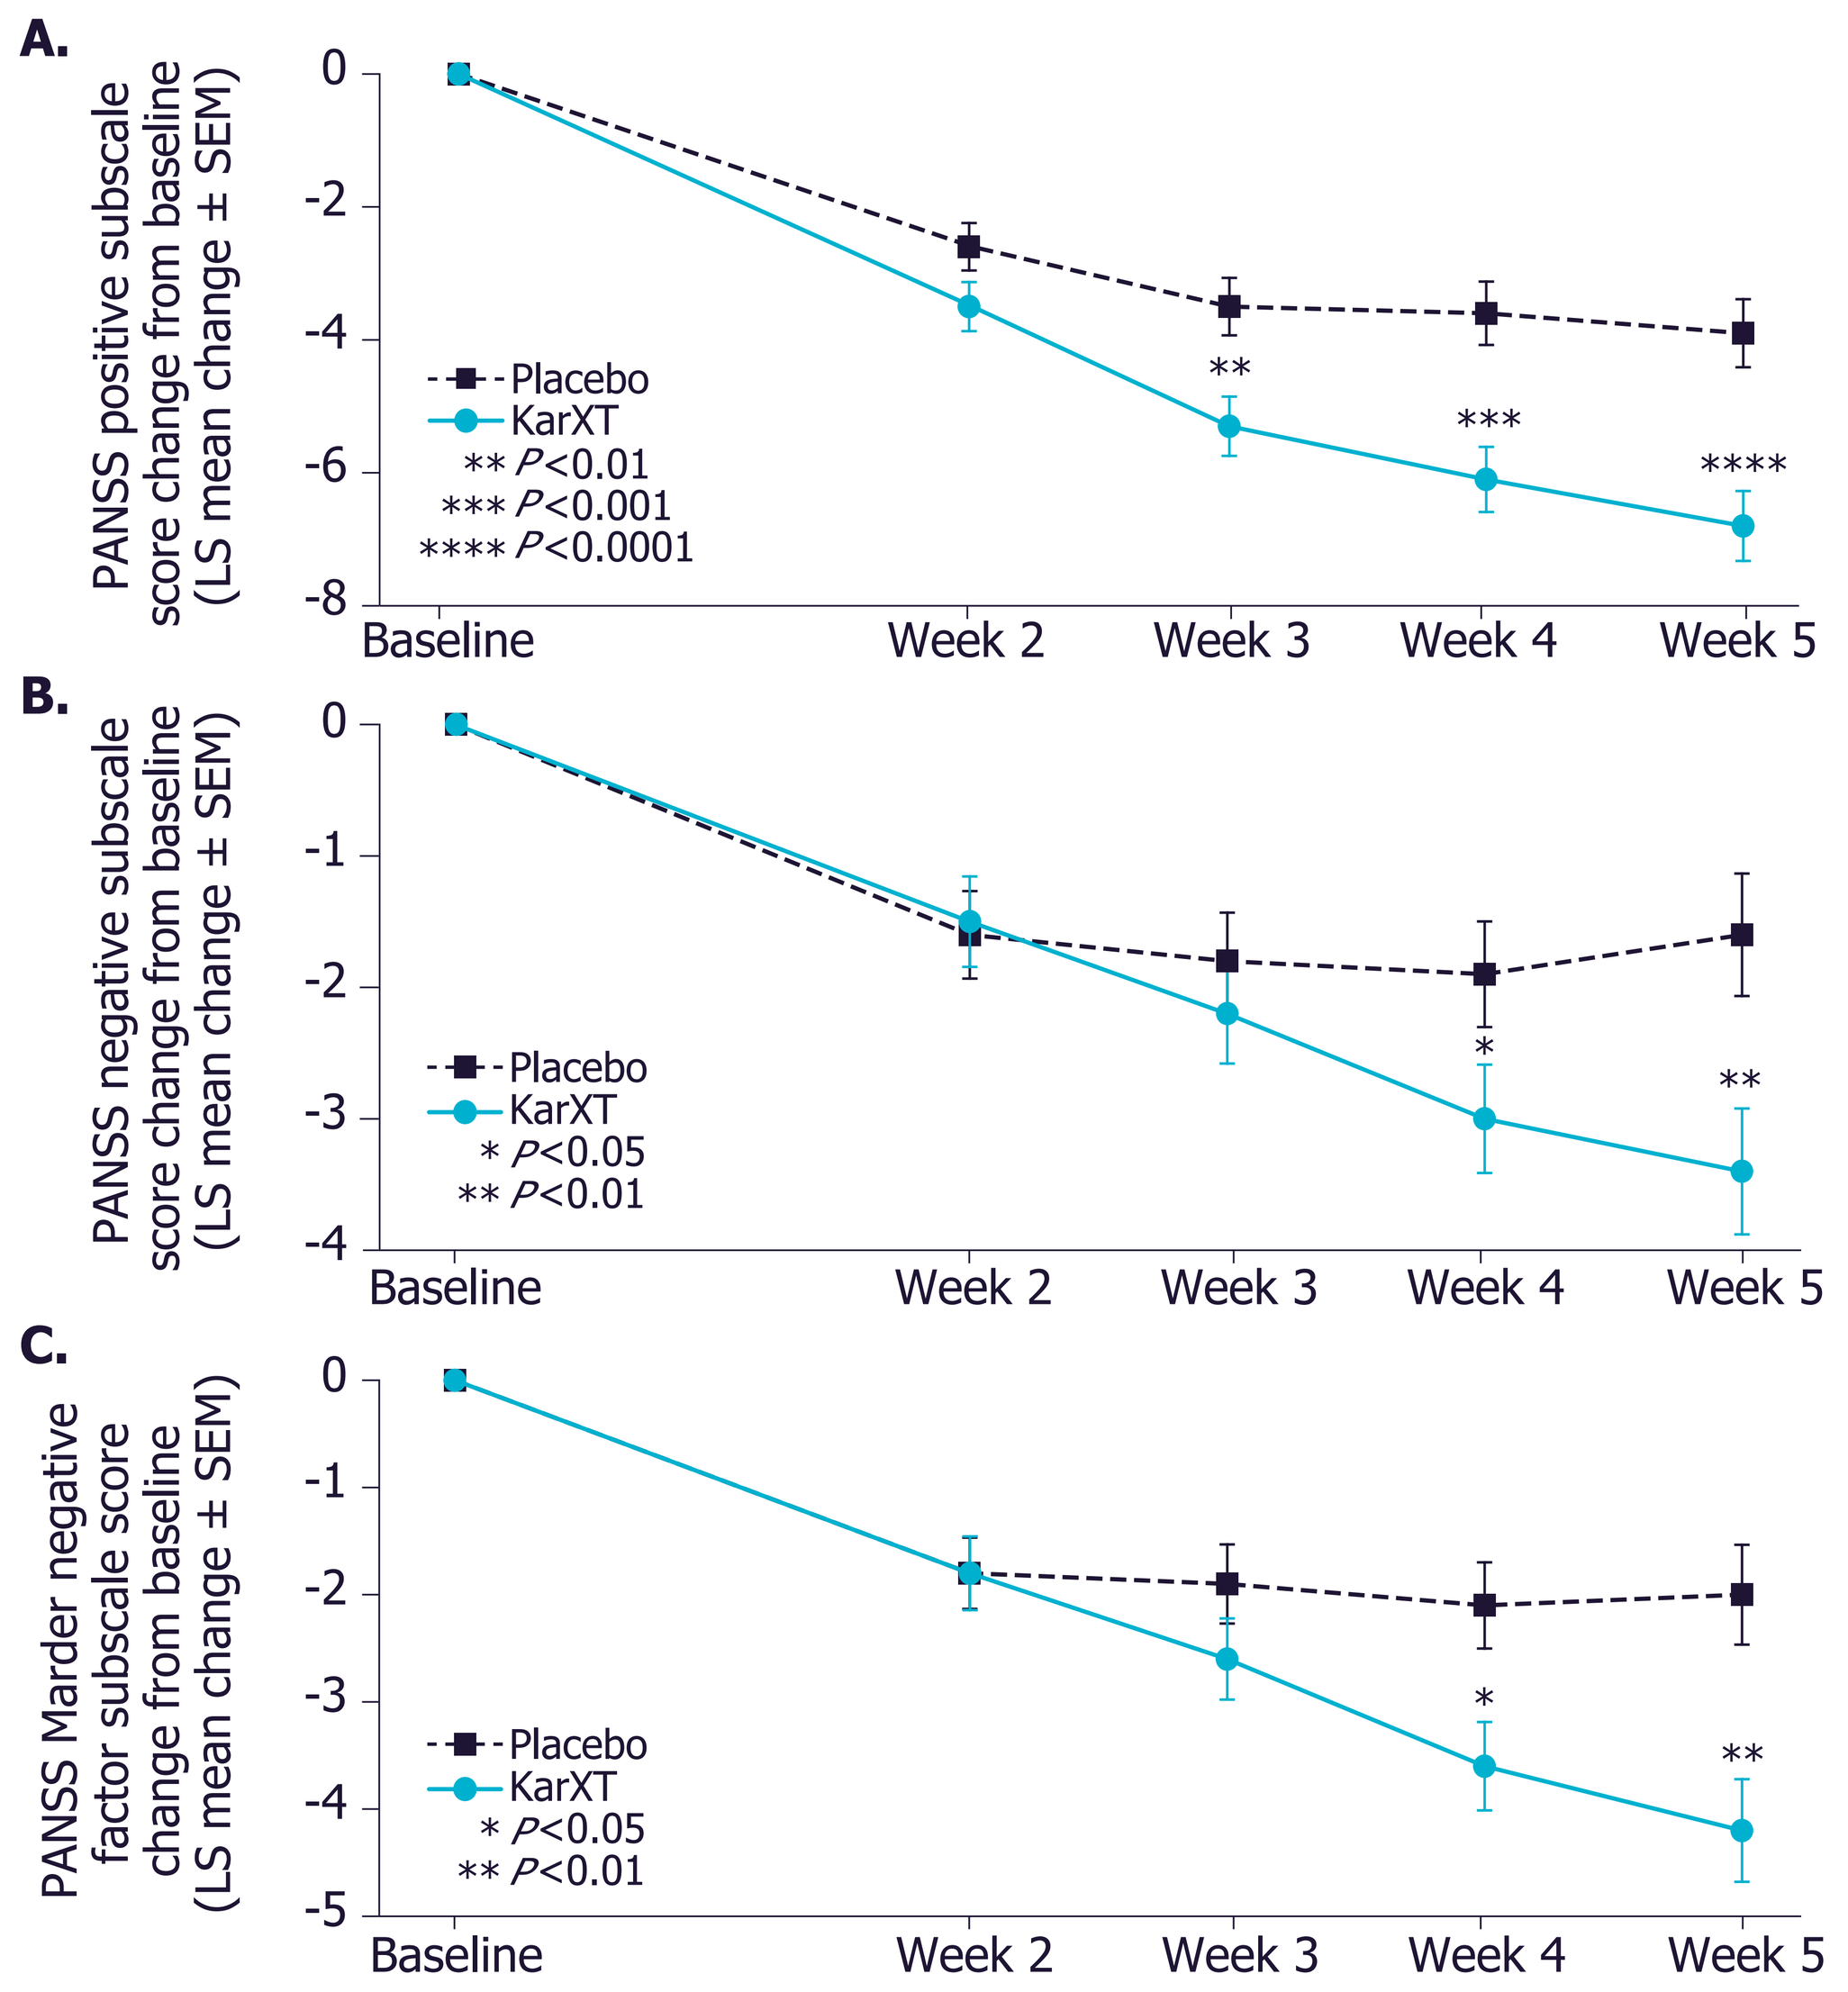
A.
PANSS positive subscale score change from baseline (LS mean change ± SEM)
0
-2
-4
-6
-8
Placebo
KarXT
** P<0.01
*** P<0.001
**** P<0.0001
Baseline
Week 2
Week 3
Week 4
Week 5
B.
PANSS negative subscale score change from baseline (LS mean change ± SEM)
0
-1
-2
-3
-4
Placebo
KarXT
* P<0.05
** P<0.01
Baseline
Week 2
Week 3
Week 4
Week 5
C.
PANSS Marder negative factor subscale score change from baseline (LS mean change ± SEM)
0
-1
-2
-3
-4
-5
Placebo
KarXT
* P<0.05
** P<0.01
Baseline
Week 2
Week 3
Week 4
Week 5

LS, least squares; PANSS, Positive and Negative Syndrome Scale; SEM, standard error of the mean.

Table 2. Safety and Tolerability During the 5-Week Treatment Period (Safety Population)

Variable	KarXT (n=126)	Placebo (n=125)
Any TEAE, n (%)	95 (75.4)	73 (58.4)
Serious TEAE,[a] n (%)	2 (1.6)	2 (1.6)
TEAE leading to discontinuation, n (%)	9 (7.1)	7 (5.6)
TEAE occurring in ≥5% of patients in the KarXT group, n (%)		
Constipation	27 (21.4)	13 (10.4)
Dyspepsia	24 (19.0)	10 (8.0)
Nausea	24 (19.0)	7 (5.6)
Vomiting	18 (14.3)	1 (0.8)
Headache	17 (13.5)	15 (12.0)
Hypertension[b]	12 (9.5)	1 (0.8)
Dizziness	11 (8.7)	4 (3.2)
Gastroesophageal reflux disease	8 (6.3)	0 (0)
Abdominal discomfort	7 (5.6)	4 (3.2)
Diarrhoea	7 (5.6)	4 (3.2)
Mean change from baseline to week 5 in body weight, kg ± SD	2.2 ± 3.62	2.0 ± 4.0
≥7% increase in body weight from baseline, n (%)	16 (12.7)	19 (15.2)

[a]Serious TEAEs were 2 cases of suicidal ideation in the KarXT group, 1 case of appendicitis in the placebo group, and 1 case of worsening of schizophrenia in the placebo group.
[b]Hypertension is the MedDRA preferred term and is not necessarily reflective of clinical hypertension.
MedDRA, Medical Dictionary for Regulatory Activities; TEAE, treatment-emergent adverse event.

- Commonly reported cholinergic TEAEs generally began within the first 2 weeks of treatment (**Figure 4**) and were intermittent and time limited in nature
 - Median number of days on treatment[b] (KarXT | placebo): vomiting (2.5 | 13.0), nausea (8.5 | 2.0), constipation (9.0 | 5.0), and dyspepsia (14.0 | 11.0)
 - Vomiting was intermittent and generally mild. About one-third of vomiting TEAEs were only a single episode of emesis
- Mean blood pressure measures were similar between KarXT and placebo at each time point throughout the trial (**Figure 5**)
 - TEAEs of hypertension were largely transient and resolved during the trial
 - Mean blood pressure at week 5 was similar to baseline in patients with a TEAE of hypertension
 - One subject in both the KarXT and placebo arms had an increase in supine systolic blood pressure of ≥15 mmHg or diastolic blood pressure of ≥10 mmHg at day 35

[b]Represents the number of days (drug | placebo) in which the patient experienced a TEAE.

Figure 4. Onset of (A) Constipation/Dyspepsia and (B) Nausea/Vomiting

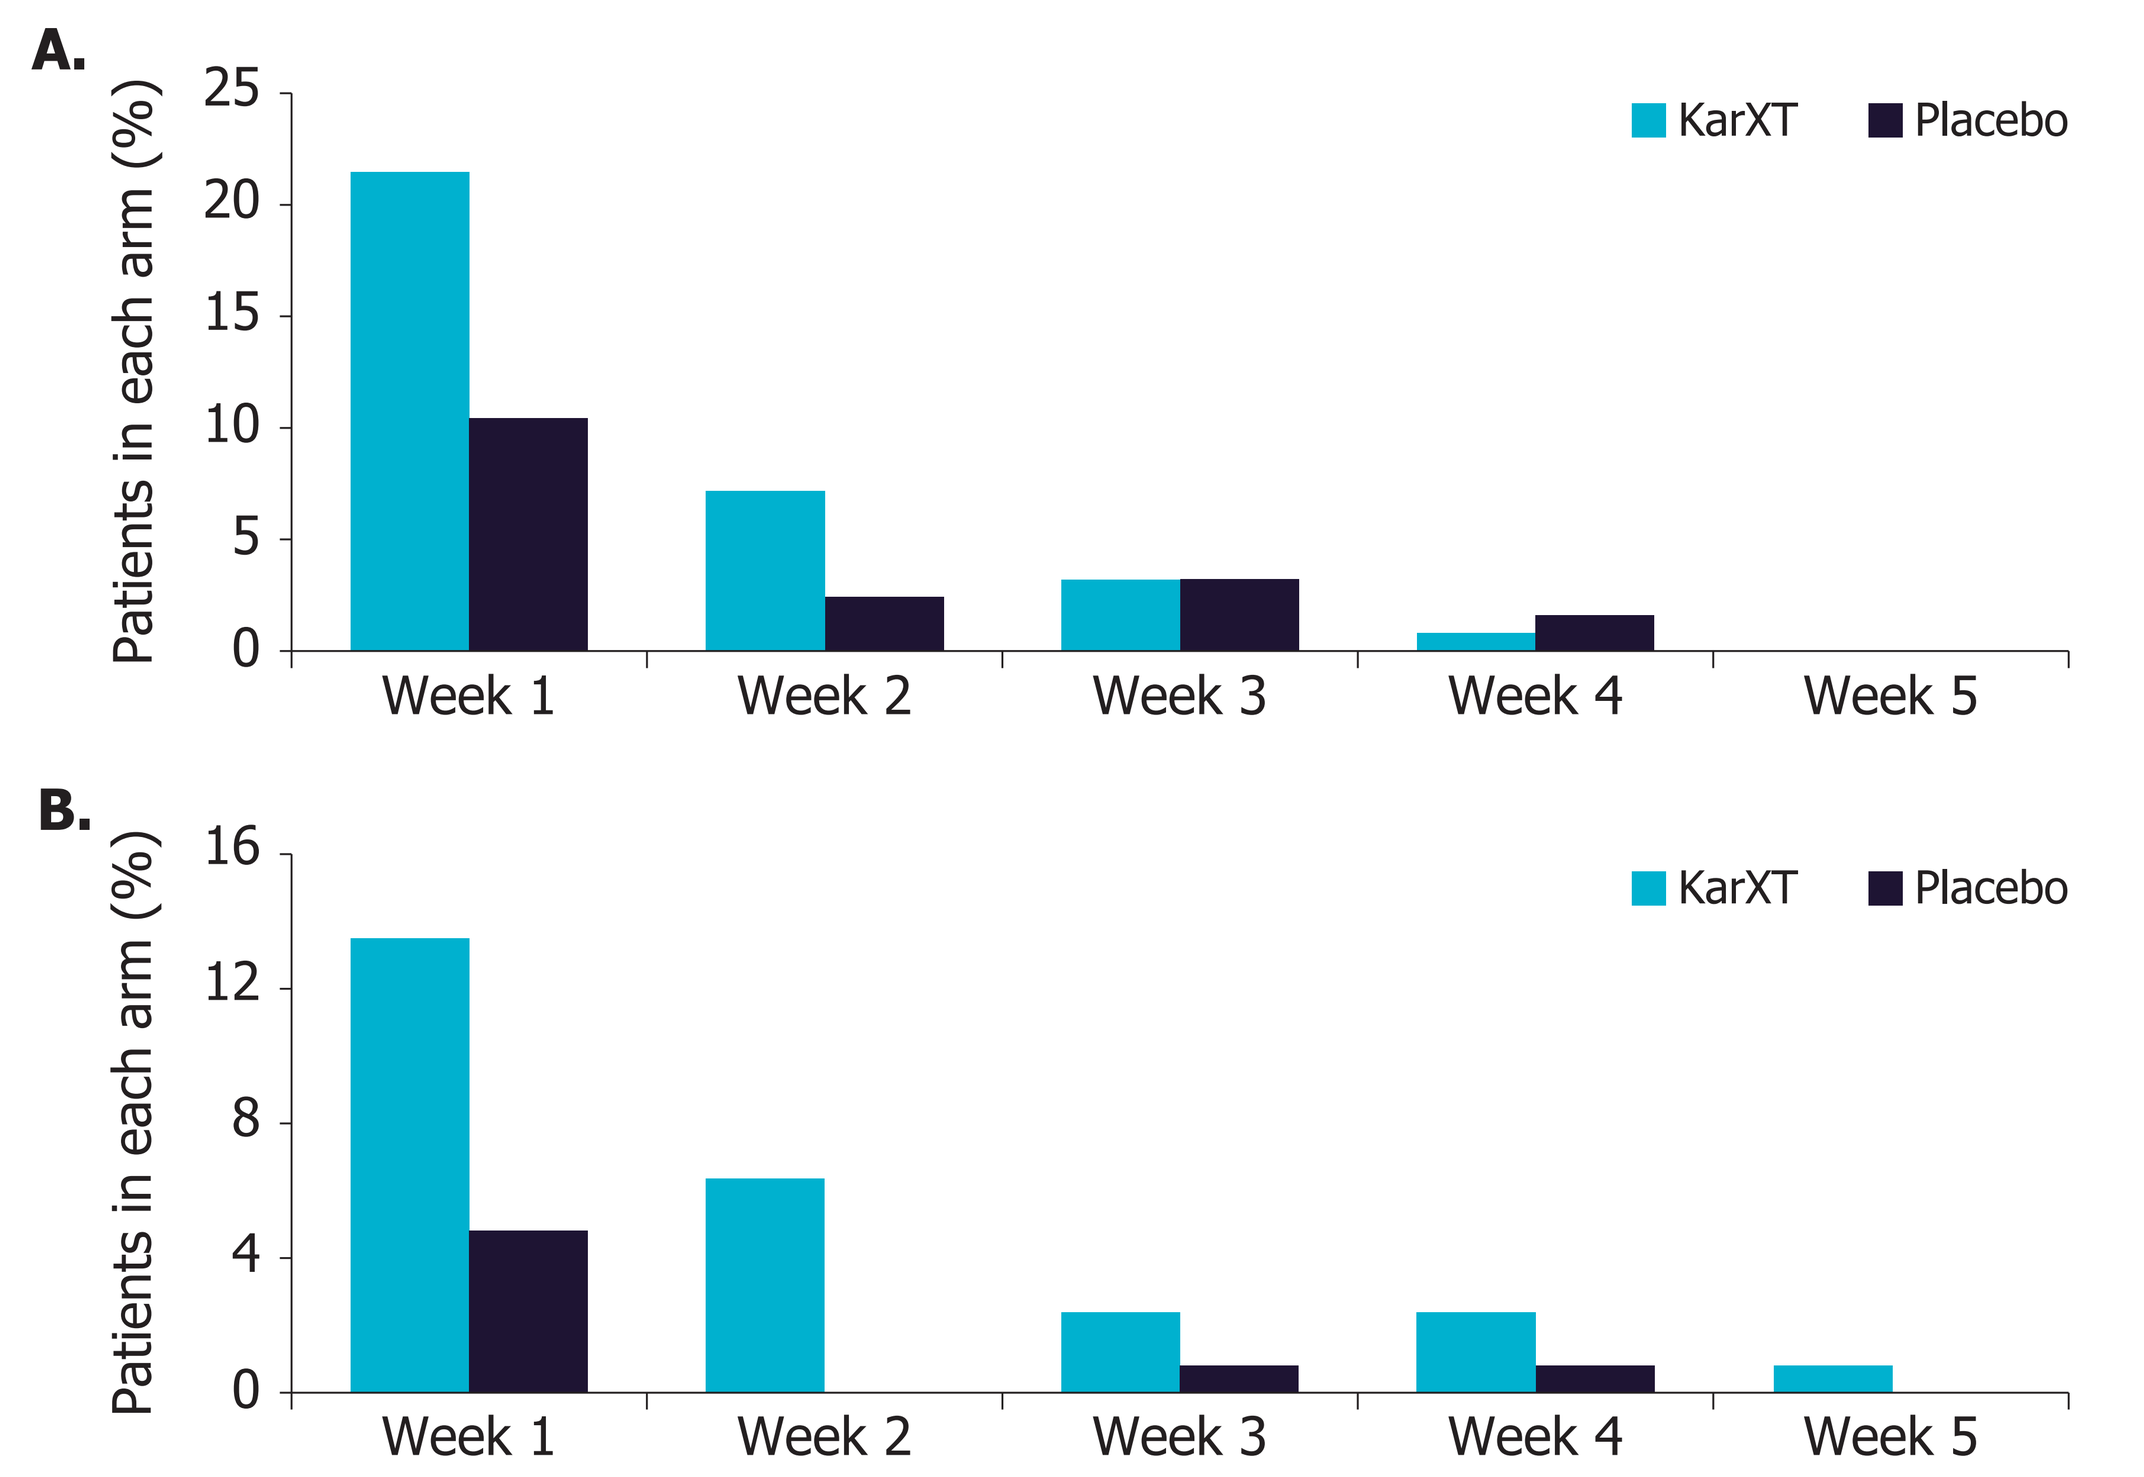
A.
Patients in each arm (%)
25
20
15
10
5
0
KarXT
Placebo
Week 1
Week 2
Week 3
Week 4
Week 5
B.
Patients in each arm (%)
16
12
8
4
0
KarXT
Placebo
Week 1
Week 2
Week 3
Week 4
Week 5

Figure 5. Observed Supine Systolic and Diastolic Blood Pressure

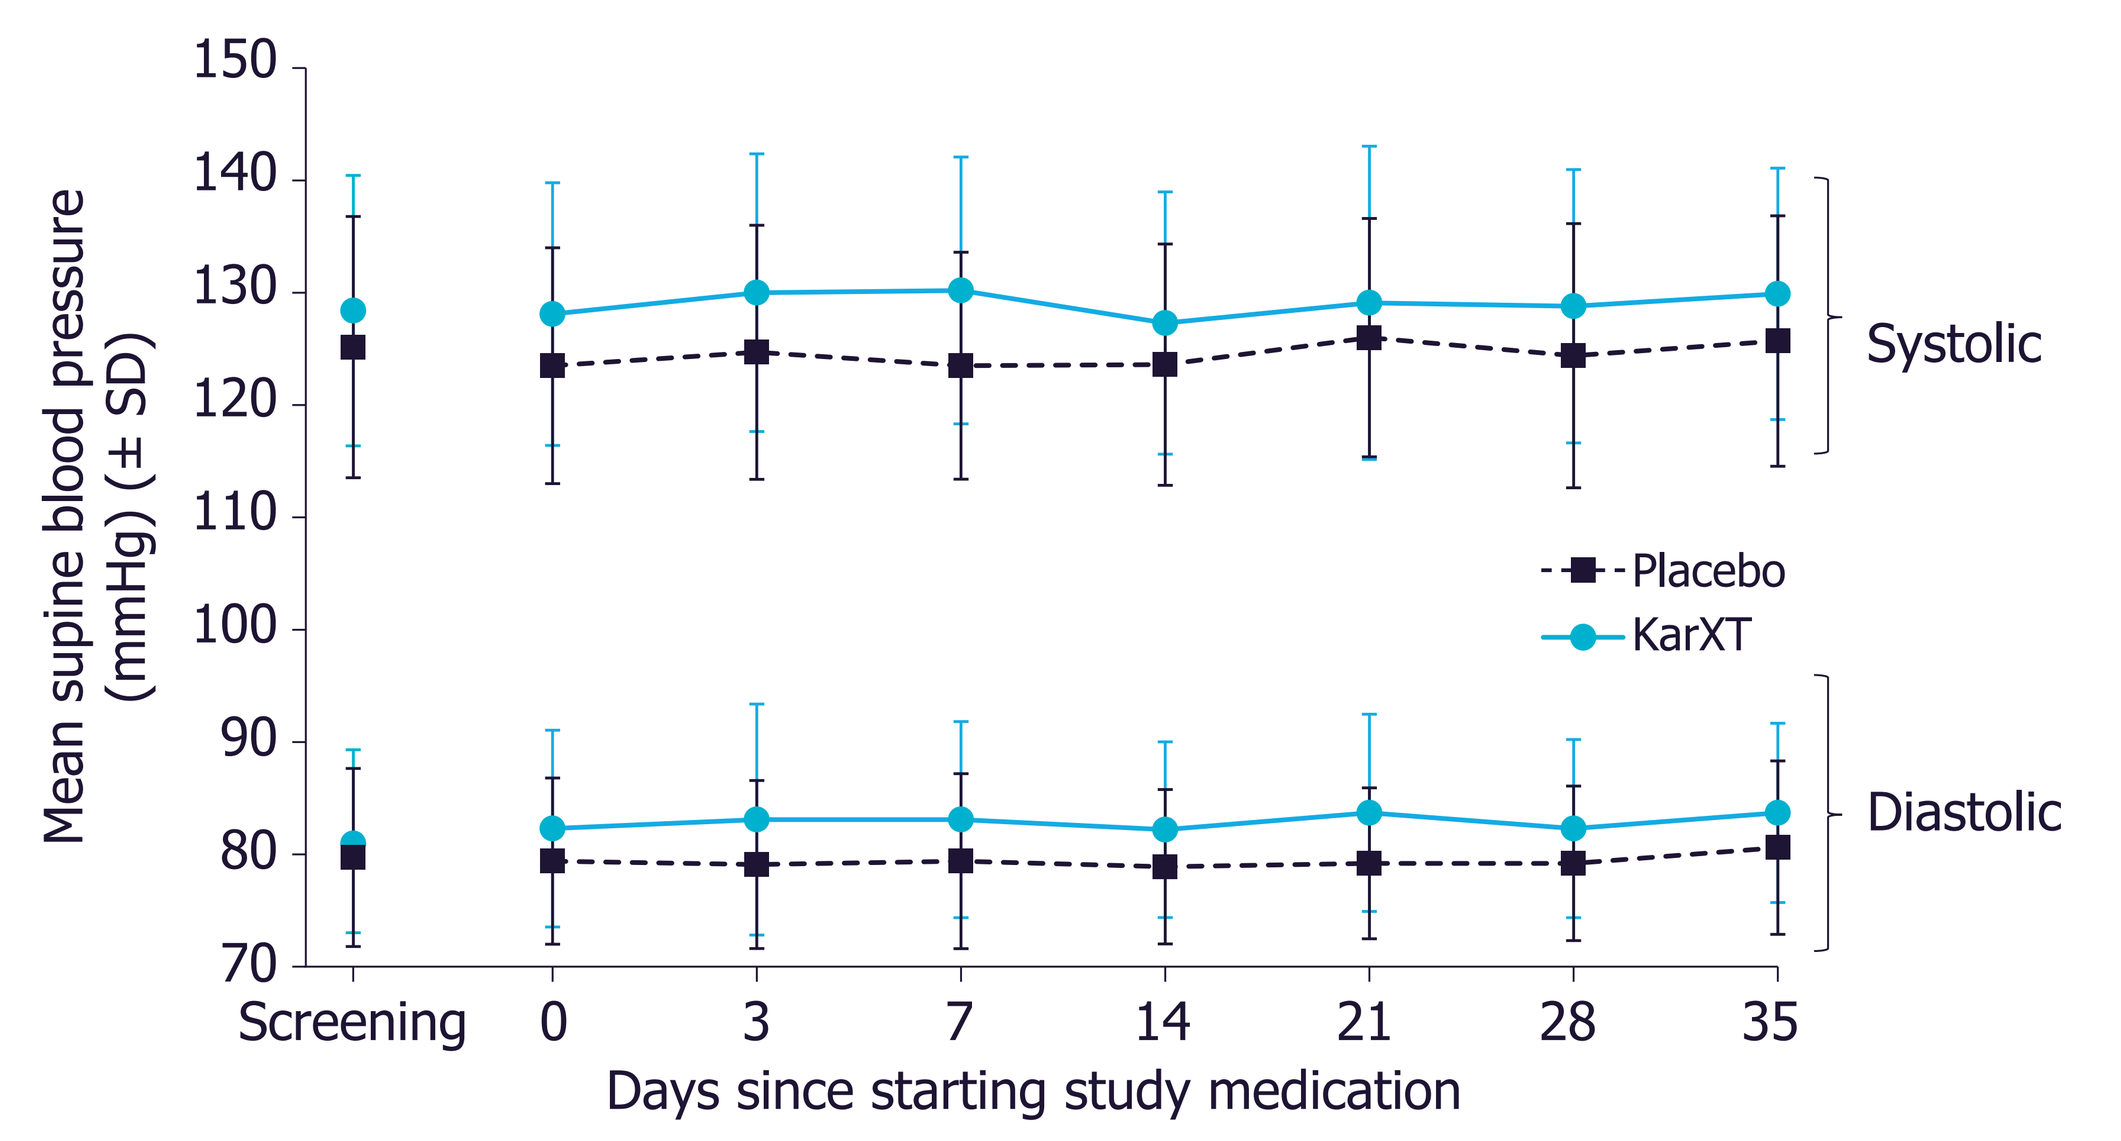
Mean supine blood pressure (mmHg) (± SD)
150
140
130
120
110
100
90
80
70
Systolic
Placebo
KarXT
Diastolic
Screening
0
3
7
14
21
28
35
Days since starting study medication

During treatment, beginning on day 0, blood pressure was measured 2 (± 1) hours after morning dose of study treatment.

Conclusions

- In the phase 3 EMERGENT-2 study, KarXT demonstrated a statistically significant and clinically meaningful improvement in PANSS total score vs placebo starting at week 2, which was maintained through all time points in the trial
- KarXT also met tested secondary endpoints, demonstrating a statistically significant reduction in both positive and negative symptoms of schizophrenia vs placebo
- Consistent with prior trials, KarXT was generally well tolerated. The most common TEAEs were all mild to moderate in severity and mostly cholinergic in nature
- KarXT was not associated with common problematic side effects of currently available antipsychotics, including somnolence, weight gain, or extrapyramidal symptoms
- KarXT has the potential to be the first in a new class of treatments for patients with schizophrenia based on muscarinic receptor agonism

Reference

1. Brannan SK, et al. *N Engl J Med.* 2021;384(8):717-726.

Disclosures

CUC has been a consultant and/or advisor to or has received honoraria from AbbVie, Acadia, Alkermes, Allergan, Angelini, Aristo Pharma, Boehringer Ingelheim, Cardio Diagnostics, Cerevel, CNX Therapeutics, Compass Pathways, Darnitsa, Gedeon Richter, Hikma, Holmusk, Intra-Cellular Therapies, Janssen/Johnson & Johnson, Karuna Therapeutics, LB Pharma, Lundbeck, MedAvante-ProPhase, MedinCell, Merck, Mindpax, Mitsubishi Tanabe Pharma, Mylan, Neurocrine Biosciences, Newron, Noven, Otsuka, Pharmabrain, PPD, Recordati, Relmada, Reviva, ROVI, Seqirus, SK Life Science, Sunovion, Sun Pharma, Supernus, Takeda, Teva, and Viatris; provided expert testimony for Janssen and Otsuka; served on a data safety monitoring board for Lundbeck, Relmada, Reviva, ROVI, Supernus, and Teva; has received grant support from Janssen and Takeda; received royalties from UpToDate; and is a stock option holder of Cardio Diagnostics, Mindpax, and LB Pharma. ASA and SKB are employees of and hold equity in Karuna Therapeutics.

Acknowledgements

The authors thank Matthew Jacobson, CMPP, and Kimberly Church of Apollo Medical Communications for medical writing and editorial support, which was funded by Karuna Therapeutics. The EMERGENT-2 study was sponsored by Karuna Therapeutics.

Copies of this poster obtained by QR code are for personal use only and may not be reused as a whole or in part without permission from the authors and study sponsor.

Presented at the 35th ECNP Congress, 15-18 October 2022, Vienna, Austria